A Phase 1a Study of AT-527, a Novel Pan-Genotypic Purine Nucleotide Prodrug Inhibitor of Hepatitis C Virus (HCV)

Xiao-Jian Zhou[1], Frédéric Vanhoutte[2], Elina Berliba[3], Pieter-Jan Berghmans[2], Maxim Bogus[3],
Steven S. Good[1], Adel Moussa[1], Keith Pietropaolo[1], Robert L. Murphy[1,4] and Jean-Pierre Sommadossi[1]

[1]Atea Pharmaceuticals, Inc., Boston, MA, USA, [2]SGS Life Sciences, Antwerp, Belgium,
[3]ARENSIA Exploratory Medicine, Republican Clinical Hospital, Chisinau, Moldova and [4]Northwestern University, Chicago, IL, USA

Background

Despite the high burden of hepatitis C virus (HCV) infection globally, there remains only a single nucleoside analog (sofosbuvir, SOF) available for use in combination with other direct-acting antivirals. The duration of SOF-containing regimens is 12 weeks for most populations. AT-527 is the salt form of a novel purine nucleotide analog prodrug in clinical development for the treatment of chronic HCV infection. The active triphosphate (TP) metabolite of AT-527 is a potent and specific inhibitor of the HCV NS5B polymerase (1,2). Unique structure modifications have conferred AT-527 with pharmacologic and antiviral properties that are differentiated from other anti-HCV nucleosides. AT-511, the free base of AT-527, was > 10-fold more active than SOF in Huh-7 cells bearing the genotype (GT) 1b replicon, with an EC_{95} of ~25nM. AT-511 showed potent pan-genotypic anti-HCV activity *in vitro* with up to 14-fold greater potency than SOF against clinical isolates harboring wild-type GT 1-4 HCV. AT-511 maintains its activity against the HCV S282T variant, with ~50-fold greater potency than SOF. GLP toxicology studies provided > 200-fold safety margin at the clinical starting dose.

Methods

Part A

- Design: Single ascending dose study; randomized, placebo (pbo)-controlled and double-blinded
- Population: Healthy male and female subjects, 18-65 years old
- Objectives: Safety/tolerability and pharmacokinetics (PK)

Cohort	Population	N (active: pbo)	AT-527 Dose (free base)	Status
1a	Healthy	6:2	50 (45) mg x 1	Complete
2a	Healthy	6:2	100 (90) mg x 1	Complete
3a	Healthy	6:2	200 (180) mg x 1	Complete
4a	Healthy	6:2	400 (360) mg x 1	Complete

Part B

- Design: Single ascending dose study; open-label
- Population: Treatment-naïve, GT1 HCV-infected subjects, HCV RNA ≥ 5 log_{10} IU/mL, non-cirrhotic
- Objectives: Safety/tolerability, PK and antiviral activity

Cohort	Population	N	AT-527 Dose (free base)	Status
1b	GT1 HCV-Infected	3	100 (90) mg x 1	Complete
2b	GT1 HCV-Infected	3	300 (270) mg x 1	Complete
3b	GT1 HCV-Infected	3	400 (360) mg x 1	Ongoing

- Clinical doses above are expressed in terms of the AT-527 salt, with the approximate AT-511 free base equivalent in parenthesis.

Results

Demographics – Part A, Healthy Subjects

	50 mg or pbo (N=8)	100 mg or pbo (N=8)	200 mg or pbo (N=8)	400 mg or pbo (N=8)
Age, mean (range) (yrs)	51 (38-65)	50 (26-63)	49 (30-65)	50 (23-65)
Weight, mean (range) (kg)	71.9 (62.5-82.3)	75.8 (53.1-93.5)	68.7 (55.2-81.1)	78.1 (61.9-93.7)
Male/Female (n)	4/4	4/4	4/4	5/3
Caucasian (n)	8	8	8	8

Demographics – Part B, HCV-Infected Subjects

	100 mg (N=3)	300 mg (N=3)
Age, mean (range) (yrs)	44 (37-53)	52 (46-55)
Weight, mean (range) (kg)	63.4 (51.0-80.0)	84.7 (67.0-107.0)
Male/Female (n)	2/1	0/3
Caucasian (n)	3	3
HCV RNA, mean (log_{10} IU/mL)	6.1	5.7
Genotype 1a/1b (n)	0/3	0/3

Adverse Events in ≥ 2 Subjects (Blinded) – Part A, Healthy Subjects

	50 mg or pbo (N=8)	100 mg or pbo (N=8)	200 mg or pbo (N=8)	400 mg or pbo (N=8)
Headache	2	2	3	2
Low back pain	0	2	2	2
Diarrhea	2	0	0	0
Nausea	1	1	0	1

Adverse Events – Part B, HCV-Infected Subjects

	100 mg (N=3)	300 mg (N=3)
Headache	2	0
Urinary tract infection	1	0

- ❖ **As the study and data management activities are ongoing, active and placebo recipients were pooled within each Part A cohort to preserve the study blind.**
- ❖ **No serious adverse events or premature discontinuations; all adverse events (AEs) were mild/moderate in intensity; no dose-related patterns were evident, including laboratory parameters, vital signs and ECGs.**

PK of AT-511 (Free Base) and AT-273 (Nucleoside Metabolite of Active TP) after a Single Dose of AT-527

	Dose (mg)	C_{max} (ng/mL)	T_{max} (h)	AUC_{tot} (ng/mLxh)	$T_{1/2}$ (h)	C_{24h} (ng/mL)
Part A, Healthy Subjects						
AT-511	50	46.4±17.6	0.5 (0.5-0.5)	36.4±12.3	0.32±0.02	--
	100	156±96.3	0.5 (0.5-1.0)	167±110	0.53±0.24	--
	200	818±443	0.5 (0.5-3.0)	656±255	0.71±0.16	--
	400	1194±401	0.5 (0.5-1.0)	1108±326	0.86±0.15	--
AT-273	50	27.9±5.62	3.5 (3.0-4.0)	285±69.4	7.07±4.59	2.28±0.95
	100	56.6±14.0	4.0 (3.0-6.0)	663±242	17.7±14.7	4.45±1.87
	200	111±38.8	5.0 (3.0-6.0)	1524±497	15.9±7.95	13.7±5.09
	400	153±49.4	6.0 (4.0-8.0)	2342±598	15.6±6.37	23.5±6.31
Part B, HCV-Infected Subjects						
AT-511	100	212±32.0	0.5 (0.5-1.0)	179±54.4	0.54±0.12	--
	300	871±590	0.5 (0.5-1.0)	818±475	0.67±0.14	--
AT-273	100	50.2±15.4	6.0 (4.0-6.0)	538±103*	8.4±4.3*	3.60±0.40
	300	96.9±38.9	6.0 (3.0-6.0)	1131±273*	8.1±2.4*	10.9±3.51

Values are reported as mean ± SD, except tor T_{max} where median (range) is reported. *Based on 24-hr profile.

Mean Plasma Concentration-Time Profiles of AT-511 and AT-273

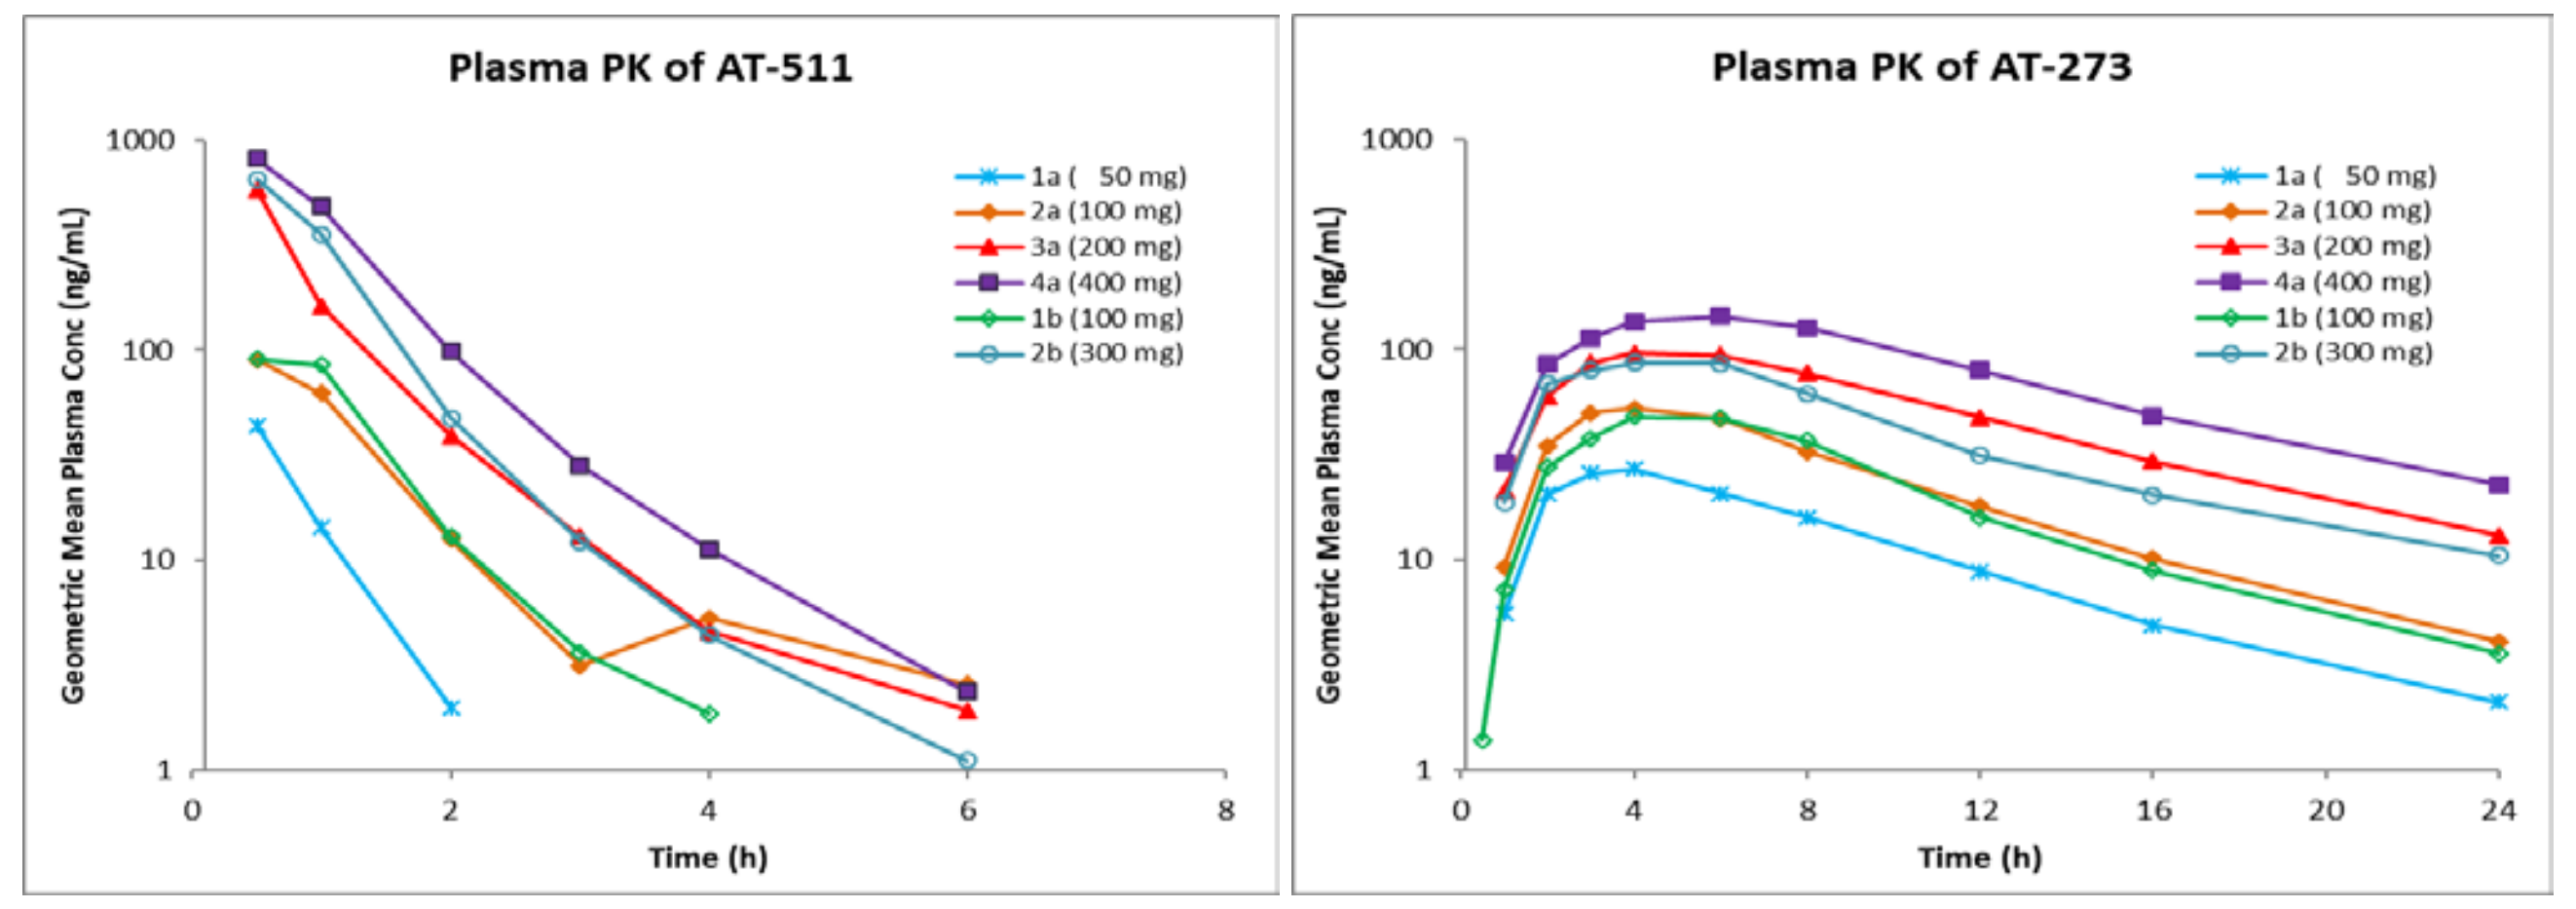
Plasma PK of AT-511
Plasma PK of AT-273
Geometric Mean Plasma Conc (ng/mL)
Time (h)
1a (50 mg)
2a (100 mg)
3a (200 mg)
4a (400 mg)
1b (100 mg)
2b (300 mg)

- ❖ **AT-511 was quickly absorbed and rapidly/extensively metabolized.**
- ❖ **AT-273, the guanosine nucleoside considered a surrogate of intracellular phosphates including the active TP, was a major metabolite and exhibited sustained plasma concentrations.**
- ❖ **Plasma exposure of AT-511 was dose-related while exposure of AT-273 was mostly dose-proportional in the studied dose range.**
- ❖ **PK was comparable in healthy subjects and HCV-infected subjects.**

Mean and Individual HCV RNA Change from Baseline After a Single Dose of AT-527

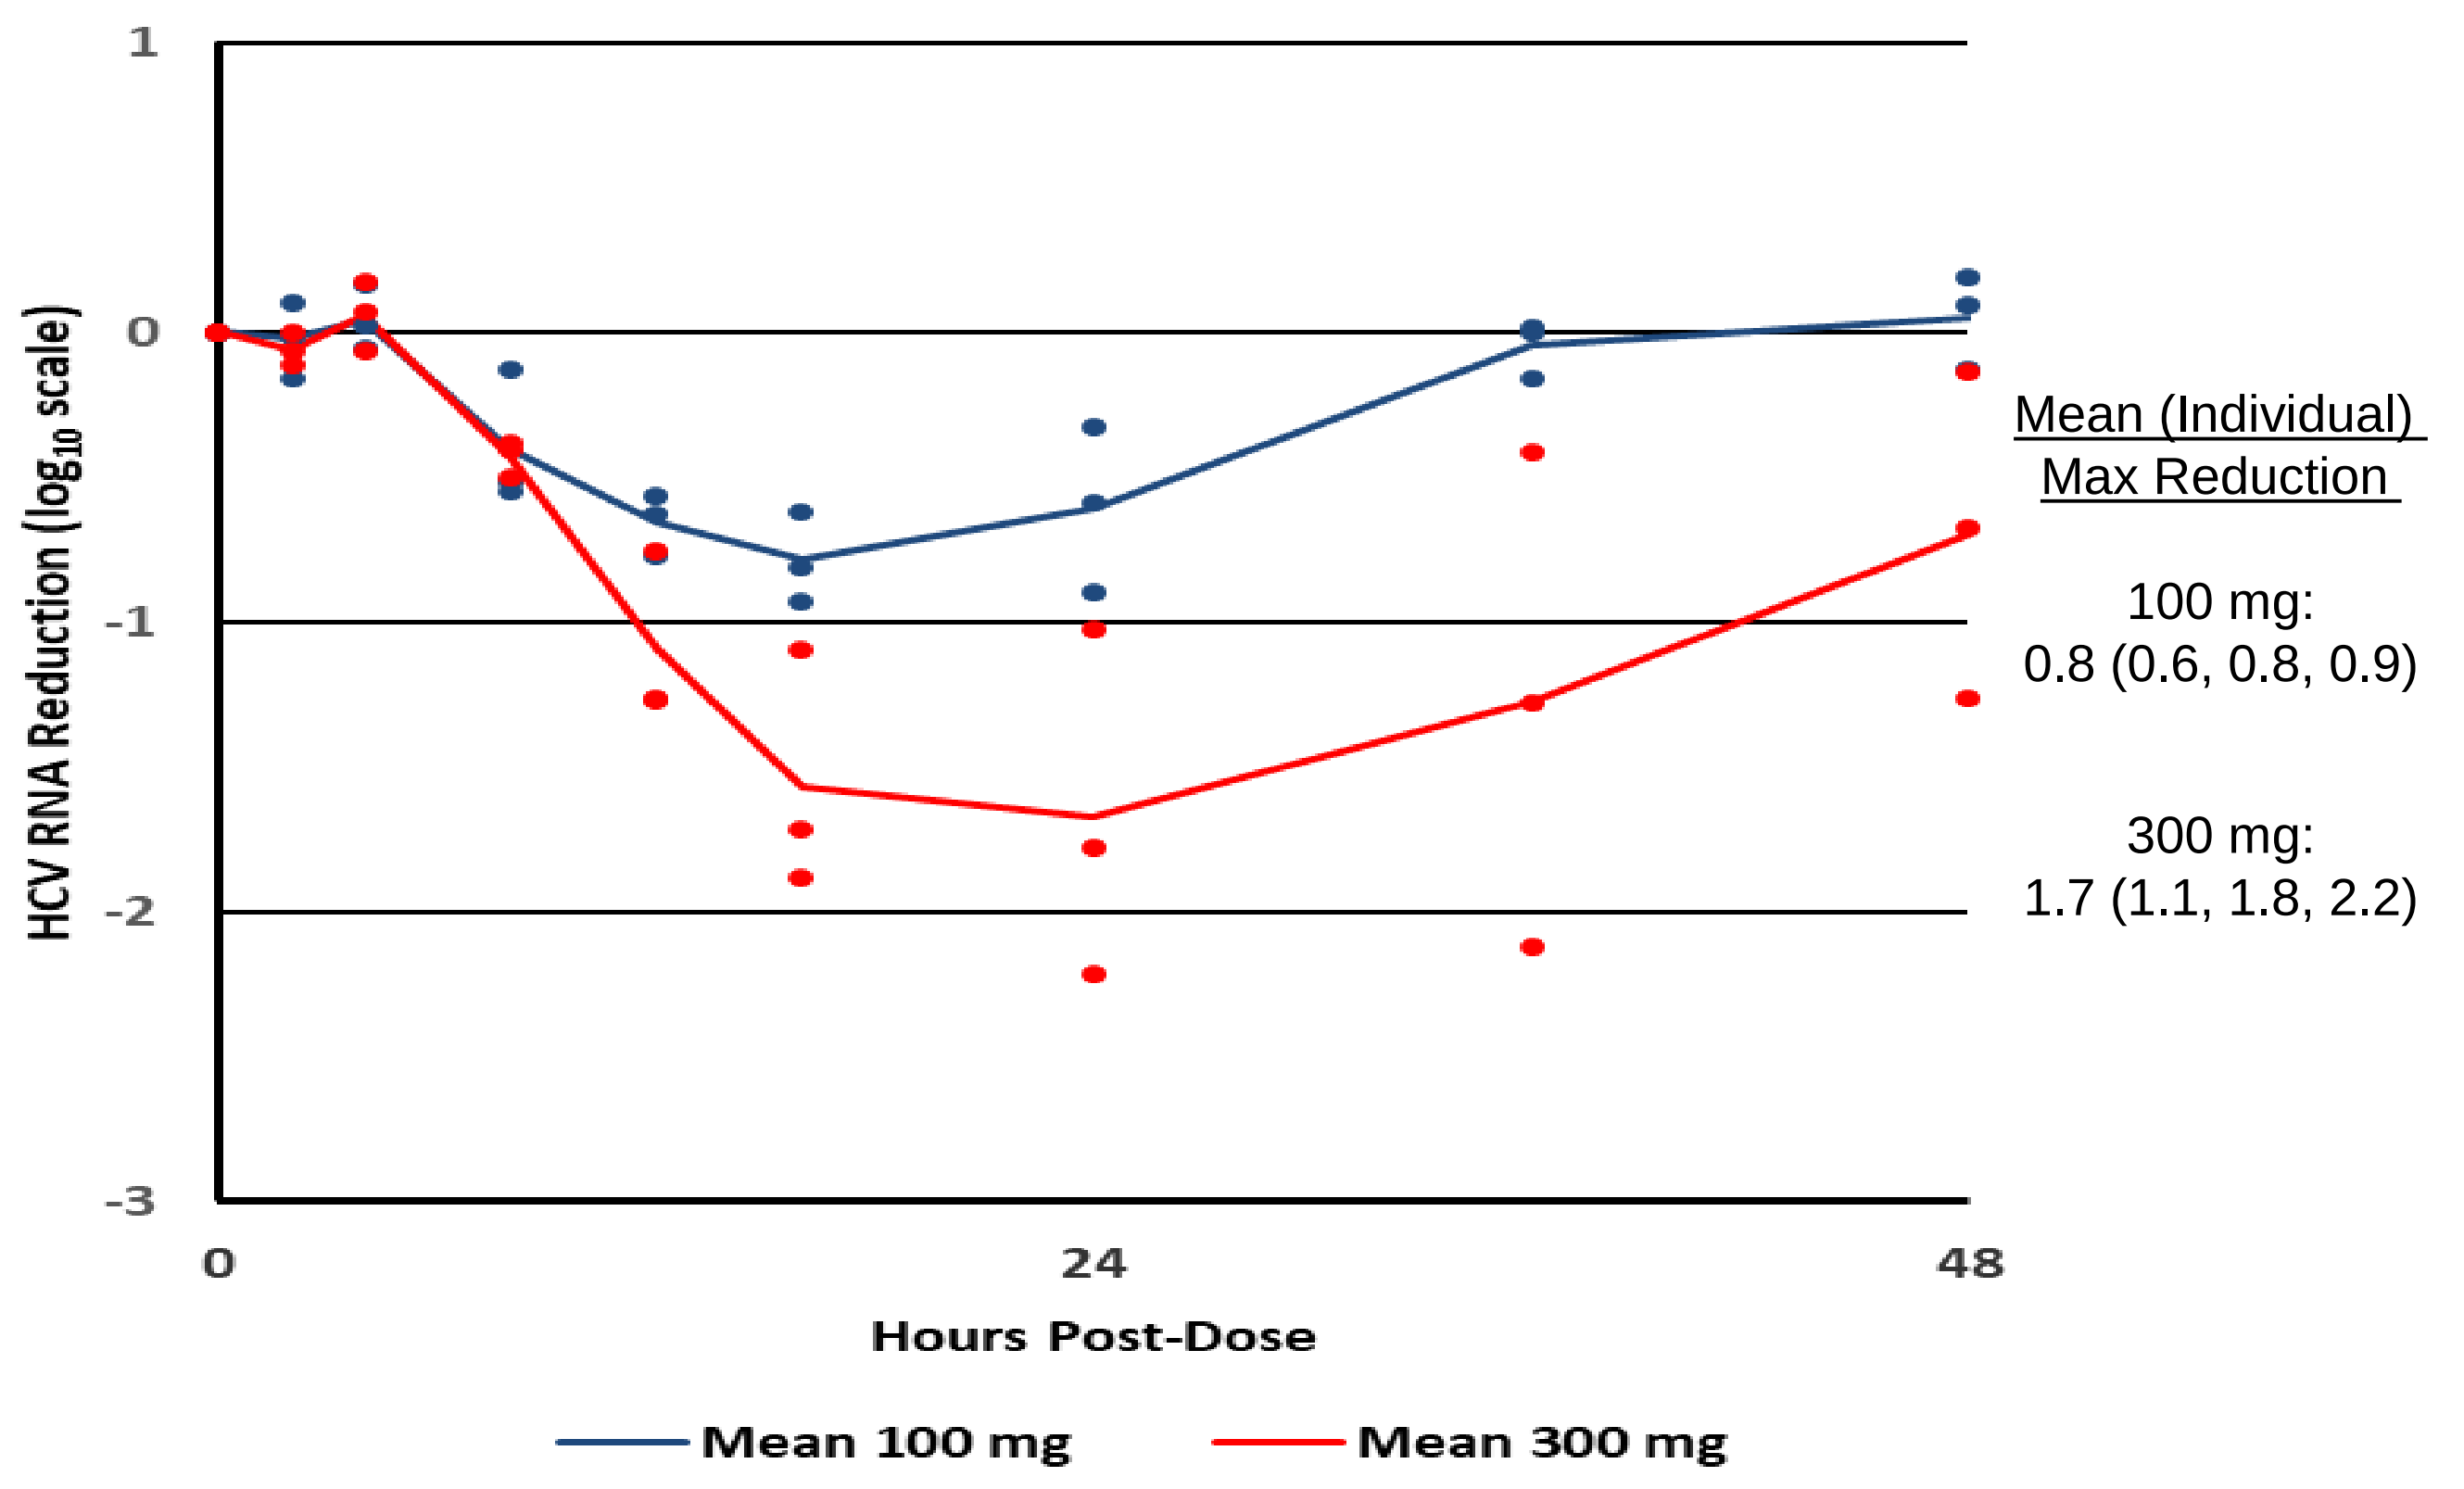
HCV RNA Reduction (log_{10} scale)
Hours Post-Dose
Mean (Individual) Max Reduction
100 mg:
0.8 (0.6, 0.8, 0.9)
300 mg:
1.7 (1.1, 1.8, 2.2)
Mean 100 mg
Mean 300 mg

- ❖ **A single 300 mg dose of AT-527 (270 mg free base equivalent) resulted in significant antiviral activity in GT1b HCV-infected subjects, with a mean maximum HCV RNA reduction of 1.7 log_{10} IU/mL.**

Individual PK/PD Analysis

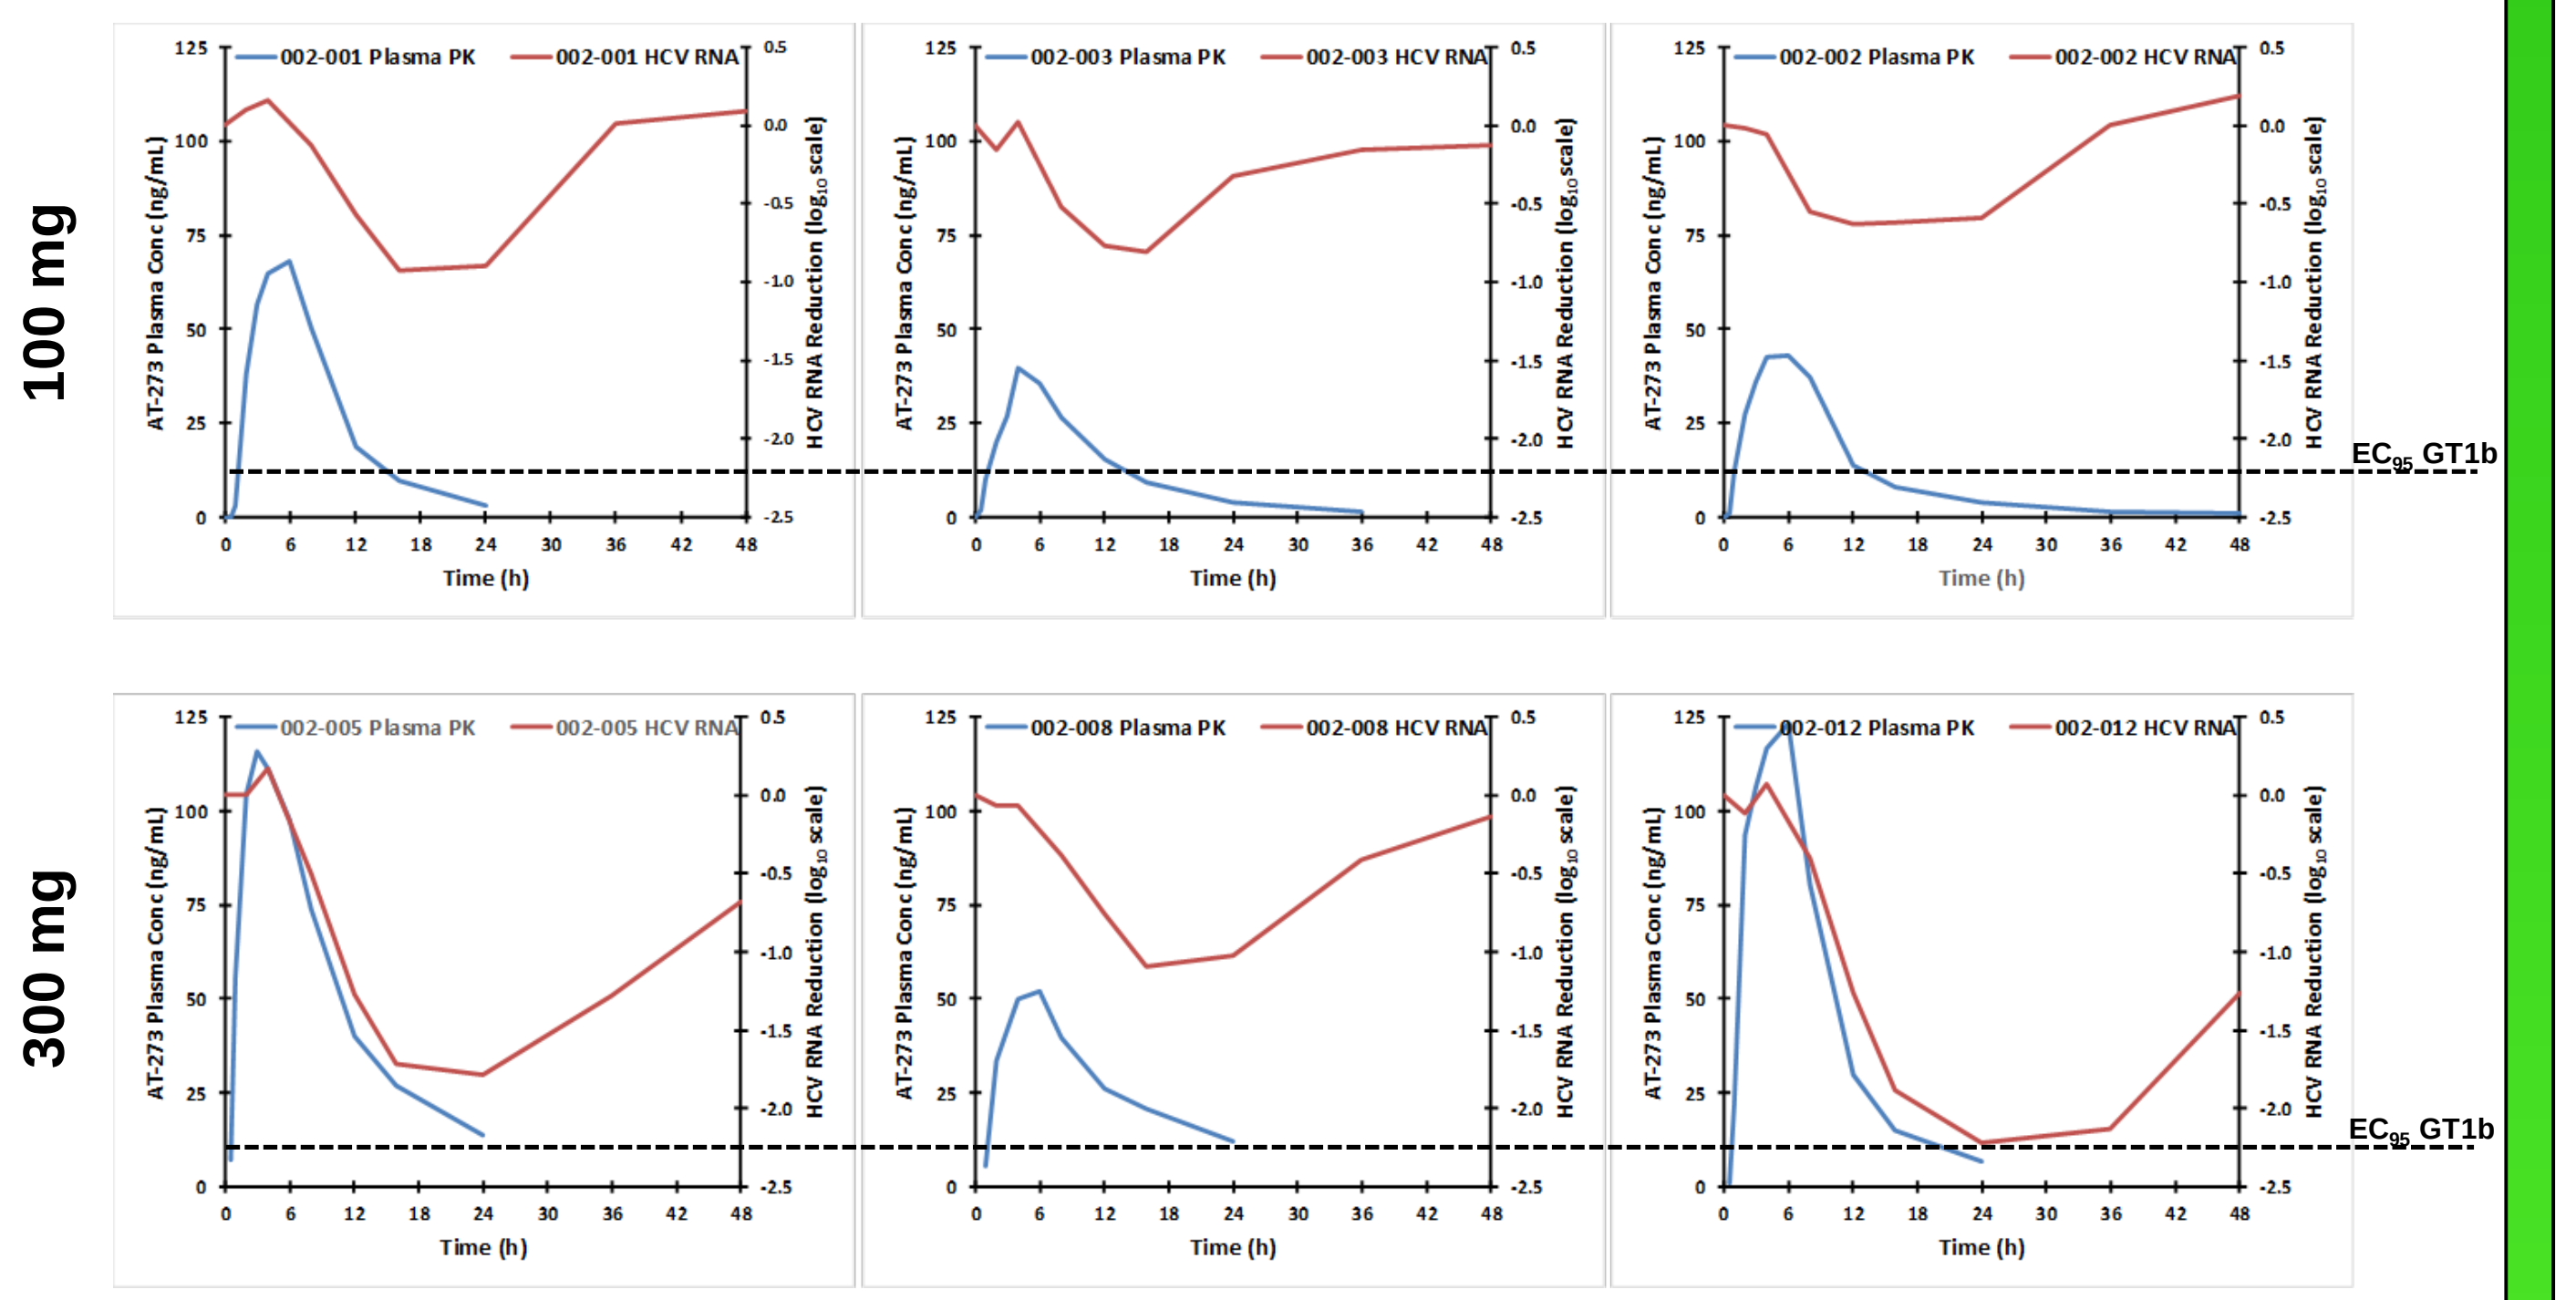
100 mg
300 mg
002-001 Plasma PK
002-001 HCV RNA
002-003 Plasma PK
002-003 HCV RNA
002-002 Plasma PK
002-002 HCV RNA
002-005 Plasma PK
002-005 HCV RNA
002-008 Plasma PK
002-008 HCV RNA
002-012 Plasma PK
002-012 HCV RNA
AT-273 Plasma Conc (ng/mL)
HCV RNA Reduction (log_{10} scale)
Time (h)
EC_{95} GT1b

- ❖ **Plasma HCV RNA reduction correlates with plasma AT-273 exposure.**
- ❖ **Viral response is sustained with AT-273 plasma concentrations > EC_{95} against GT1b.**

Predicted Maximum HCV RNA Reduction vs. AT-273 AUC Achievable after a Single Dose of AT-527 up to 600 mg

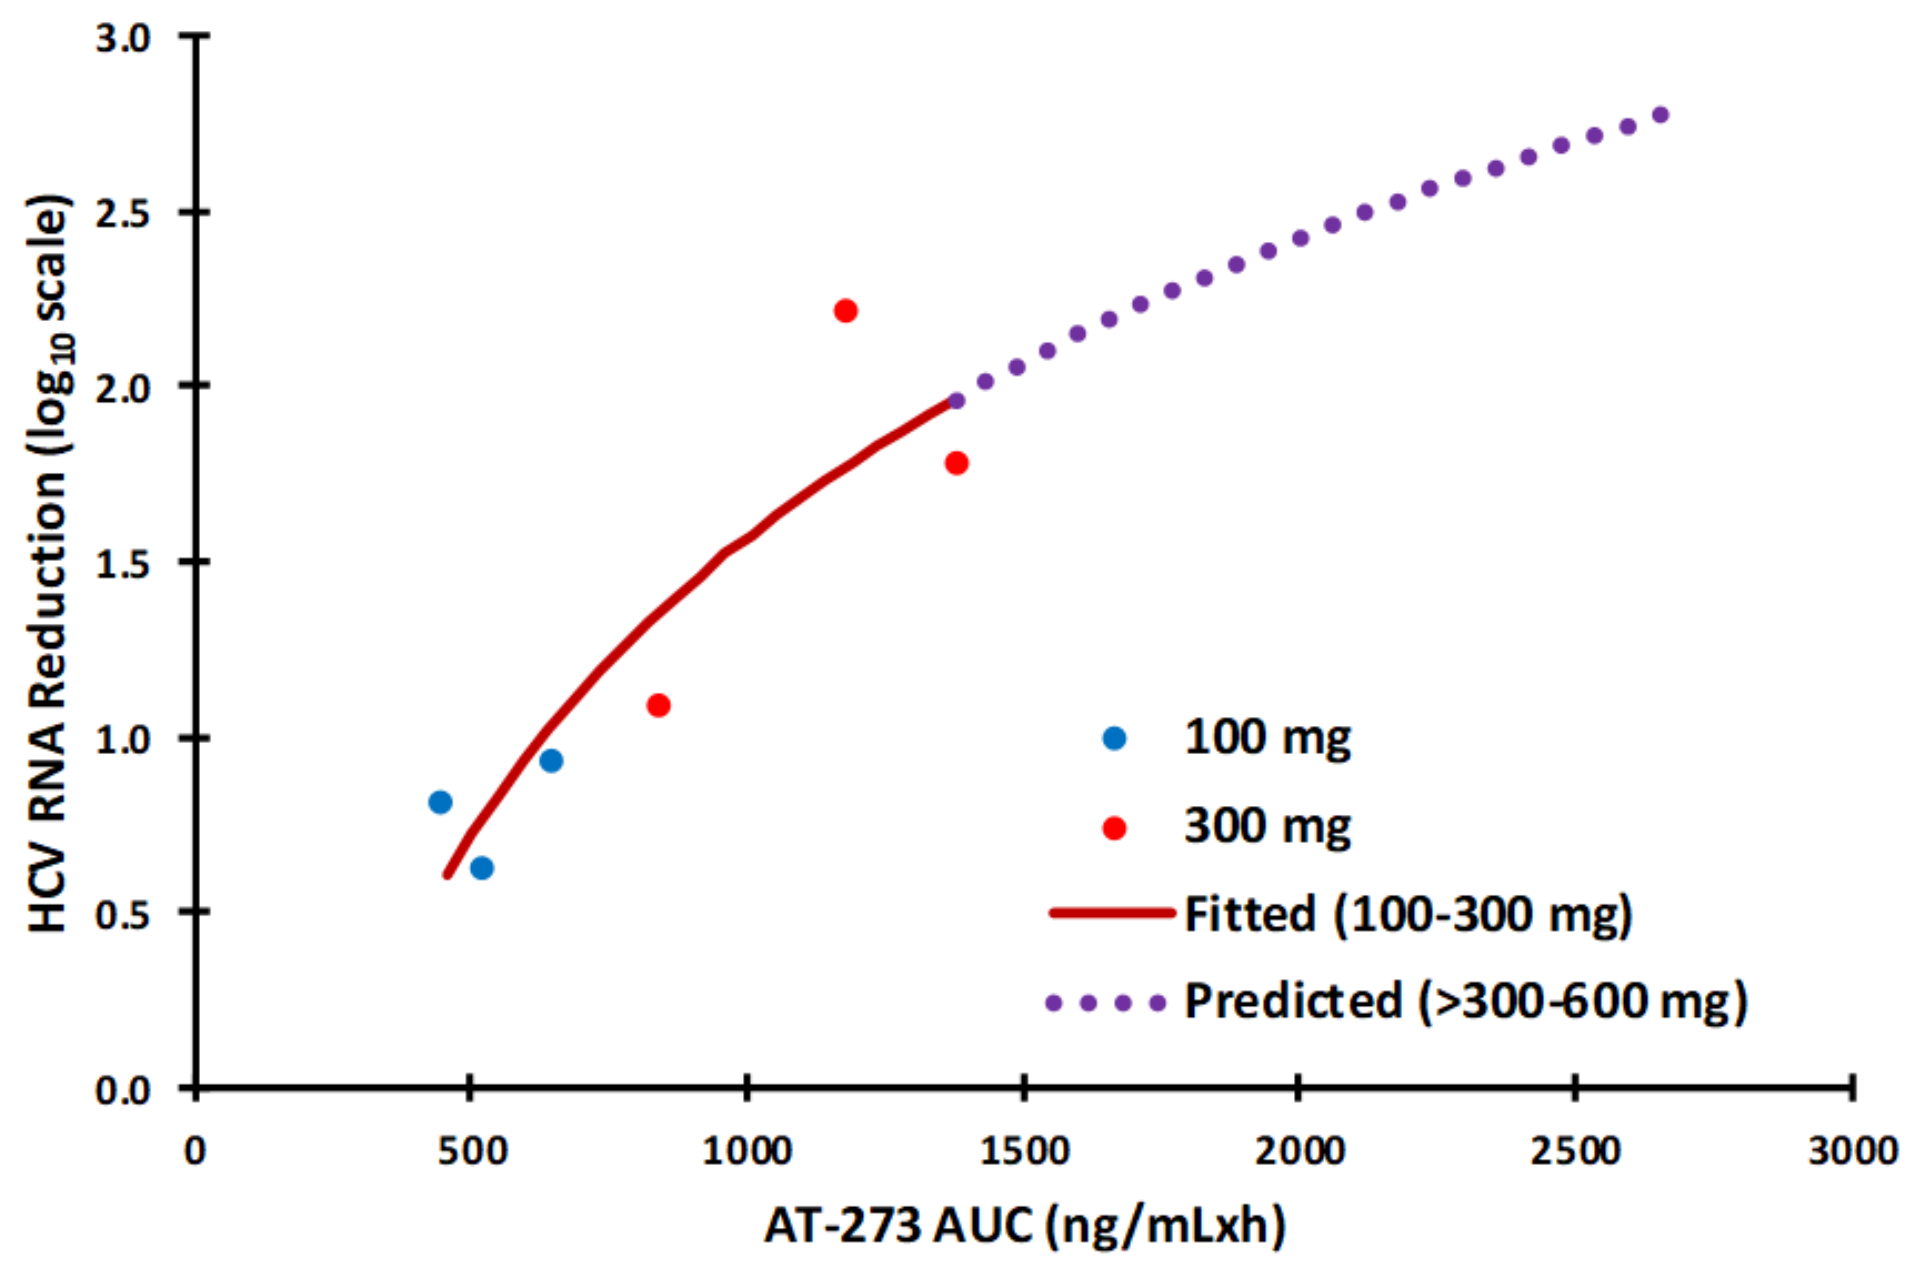
HCV RNA Reduction (log_{10} scale)
AT-273 AUC (ng/mLxh)
100 mg
300 mg
Fitted (100-300 mg)
Predicted (>300-600 mg)

Predicted Steady-State Trough Levels of AT-273 Exceed AT-511 EC_{95} against Clinical Isolates of HCV GT 1-4

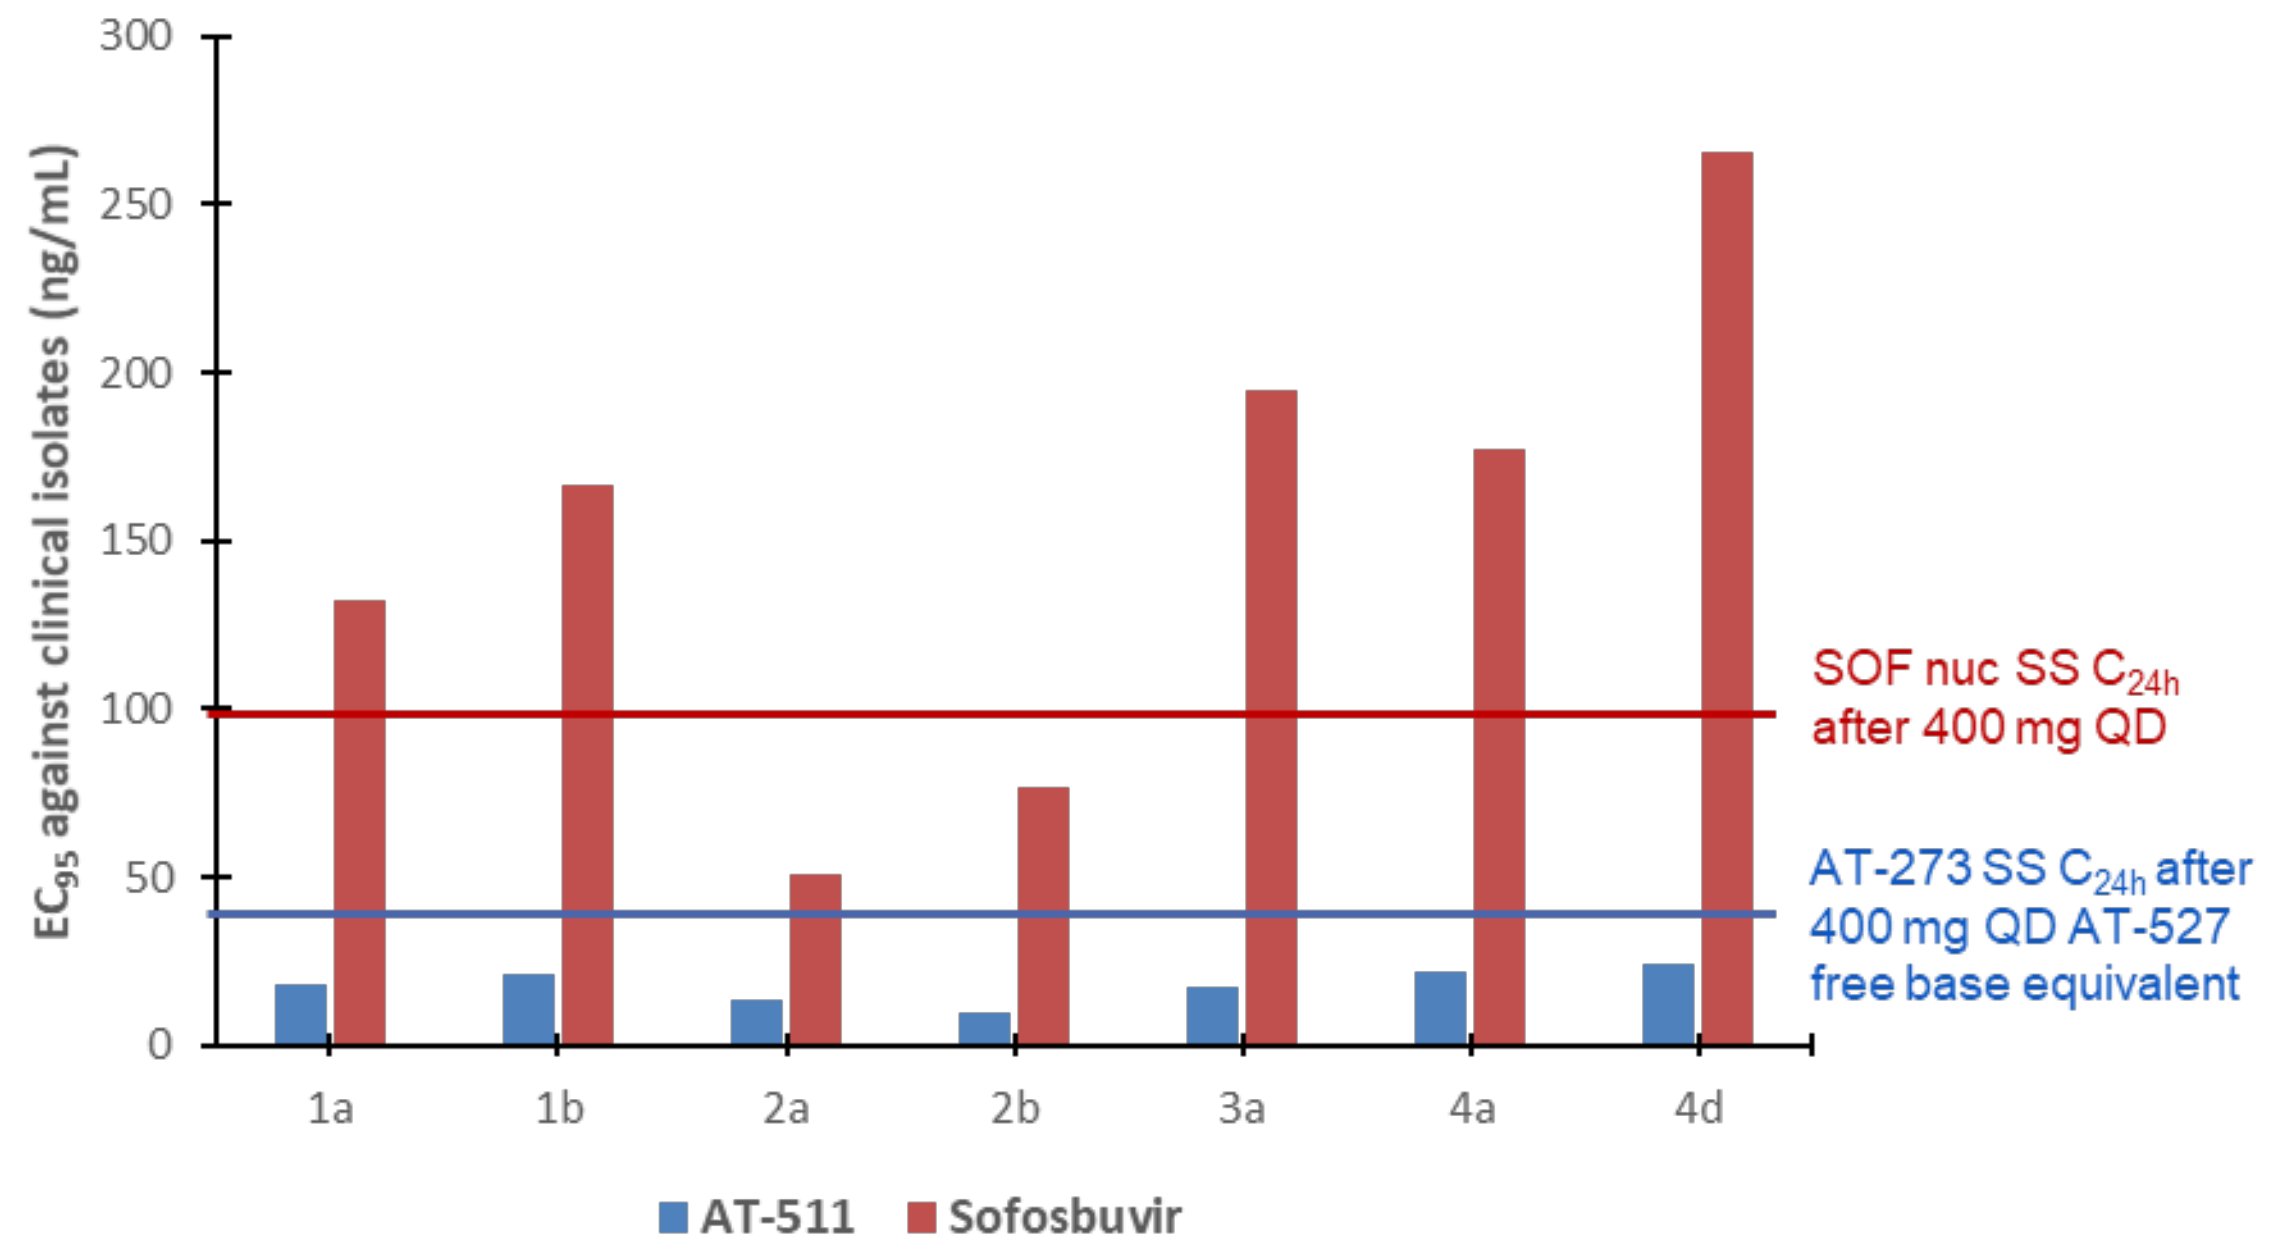
EC_{95} against clinical isolates (ng/mL)
SOF nuc SS C_{24h} after 400 mg QD
AT-273 SS C_{24h} after 400 mg QD AT-527 free base equivalent
1a
1b
2a
2b
3a
4a
4d
AT-511
Sofosbuvir

- ❖ **Predicted steady-state (SS) trough plasma levels (C_{24h}) of AT-273 after 400 mg QD free base equivalent dosing of AT-527 exceed the *in vitro* EC_{95} of AT-511 against all tested clinical isolates of GT 1-4 HCV-infected patients.**
 - **SS C_{24h} levels of SOF clinical dose (400 mg QD) only cover clinical isolates of GT2.**

Conclusions

- ➢ AT-527 was well tolerated after a single dose in both healthy and HCV-infected subjects.
- ➢ After single oral doses, plasma exposure of AT-273, the nucleoside metabolite of the active nucleotide, was mostly dose-proportional over the studied dose range.
- ➢ PK parameters were comparable in both healthy and HCV-infected subjects.
- ➢ Significant antiviral activity was observed in GT1b HCV-infected subjects after a single 300 mg dose of AT-527 (270 mg free base equivalent), with a mean maximum HCV RNA reduction of 1.7 log_{10} IU/mL.
 - This compares to ~2 log_{10} IU/mL reduction after 1 day of 400 mg SOF monotherapy in GT1a HCV-infected subjects (3).
- ➢ PK/PD analysis indicated that antiviral activity correlated with plasma exposure of AT-273, predicting a more profound viral response at higher drug exposure.
 - Predicted steady-state troughs of AT-273 after 400 mg QD AT-527 free base equivalent exceed the *in vitro* EC_{95} of AT-511 against all tested clinical isolates of GT 1-4 HCV patients, suggesting that AT-527 will have potent pan-genotypic antiviral activity.
- ➢ Higher doses are being evaluated in Part B of this ongoing study. These data support initiation of a 7-day proof-of-concept study, in which QD doses of AT-527 will be evaluated as a single agent in HCV-infected subjects.

References

1. S.S. Good et al. (2015) Discovery of AT-337, AT-339 and AT-511, Three Highly Potent and Selective Nucleotide Prodrug Inhibitors of HCV Polymerase. Abs. 2266. Hepatology. 62(1, Suppl): 1310A-11A
2. S.S. Good et al. (2016) AT-337, AT-511 and its Salt Form, AT-527: Novel Potent and Selective Pan-genotypic Purine Nucleotide Prodrug Inhibitors of HCV Polymerase. Abs. 1452. Hepatology. 64(1, Suppl): 726A
3. E. Lawitz et al. (2011) Once Daily Dual-Nucleotide Combination of PSI-938 and PSI-7977 Provides 94% HCV RNA < LOD at Day 14: First Purine/Pyrimidine Clinical Combination Data (The Nuclear Study) . Abs. 1370. Journal of Hepatology. 54(1, Suppl): S543.

Acknowledgements

The authors would like to thank the subjects who participated in this study. In addition, thanks to the staff at SGS Life Sciences and ARENSIA Exploratory Medicine.

Disclosures: XJZ, SSG, AM, KP and JPS are employees of Atea Pharmaceuticals. RLM is a consultant to Atea Pharmaceuticals.